Prevalence of Healthcare Conditions and Services Used by Patients with Myotonic Dystrophy (DM1 and DM2) Prior to Diagnosis: a Real-World Data Analysis

John W. Day[1], Kathryn A. Munoz[2], Richard A. Brook[3], Bradley McEvoy[2], Mark C. Stahl[2], Kelly DiTrapani[2], Nathan L. Kleinman[3], Chao-Yin Chen[2], Li-Jung Tai[2]

[1]Stanford University Medical Center; [2]Avidity Biosciences, Inc.; [3]Better Health Worldwide. All authors have met authorship criteria.

Objectives

Describe healthcare conditions, costs, and services received by DM patients versus matched non-DM controls (MCs) 1 year before confirmed DM diagnosis

Background

- DM is a rare, dominantly inherited, monogenic, multisystem disease that causes myotonia, progressive muscle weakness, and atrophy, along with respiratory, gastrointestinal, cardiac, and central nervous system dysfunction, which significantly impacts quality of life[1-3]
- There are two major types of DM (type 1 and type 2)[1]
- Patients experience significant physical limitations, pain, fatigue, and a negative impact on wellbeing[4,5]
- Currently, there is no cure or targeted treatment for DM[3]
- Limited literature exists to quantify the burden of disease prior to diagnosis

Methods

- Retrospective database analysis to compare conditions, costs, and services of DM patients versus those of MCs
 - Database: IQVIA US PharMetrics® Plus
 - Time frame: January 2010 through March 2021
- The DM cohort is defined as having ≥2 DM claims ≥30 days apart
 - Claims identified by International Classification of Disease Ninth Revision (ICD-9) code 359.21 and Tenth Revision (ICD-10) code G71.11, which does not differentiate between DM subtypes
- The first diagnosis date was used for the index date
- DM patients were matched to a 5% random sample of eligible non-DM controls
 - Matching was done using R's Matchit procedure, with nearest neighbor matching (exact matching on month of index date)
 - Cohorts were matched (5-MC:1-DM) on index month and baseline age, region, gender, plan, and payer type
- All subjects (patients and MCs) had a minimum of 12 months of continuous data prior to their index date
 - Because our focus was on the events preceding diagnosis, the index date was not included in the 1-year pre-index evaluation period
- Healthcare conditions, costs, and services used during the 12 months before each subject's index date were compared using 283 US Agency for Healthcare Research & Quality (AHRQ) comorbidity categories
- Comparisons were made using *t*-tests for continuous variables and chi-square tests for discrete variables
- Total per member per year (PMPY) direct medical and prescription (Rx) costs were calculated for both DM and MC cohorts
- All costs were inflation adjusted using the US Bureau of Labor Statistics Consumer Price Index (CPI) for December 2020
 - Medical claims used the medical cost CPI and prescription claims used the prescription cost CPI

Sponsorship
This poster is sponsored by Avidity Biosciences

Abbreviations
AHRQ, Agency for Healthcare Research and Quality; CPI, Consumer Price Index; DM, myotonic dystrophy; ICD, International Classification of Disease; MC, matched control; PMPY, per member per year; Rx, prescription; SD, standard deviation.

References
[1]Udd B and Krahe R. *Lancet Neurol.* 2012;11(10):891–905; [2]Gourdon G and Meola G. *Front Cell Neurosci*. 2017;11:101; [3]LoRusso S, et al. *Neurotherapeutics*. 2018;15(4):872–84; [4]Hagerman KA, et al. *Muscle Nerve*. 2019;59(4):457–64; [5]Landfeldt E, et al. J Neurol. 2019;266(4):998–1006.

Prevalence of Healthcare Conditions and Services Used by Patients with Myotonic Dystrophy (DM1 and DM2) Prior to Diagnosis: a Real-World Data Analysis

John W. Day[1], Kathryn A. Munoz[2], Richard A. Brook[3], Bradley McEvoy[2], Mark C. Stahl[2], Kelly DiTrapani[2], Nathan L. Kleinman[3], Chao-Yin Chen[2], Li-Jung Tai[2]

[1]Stanford University Medical Center; [2]Avidity Biosciences, Inc.; [3]Better Health Worldwide. All authors have met authorship criteria.

Results

- We identified 1,694 DM patients and 8,470 MCs
 - There were no significant differences between cohorts in descriptive characteristics (Table 1)
 - The cohorts had significant (*p*<0.0001) pre-index differences for the Charlson Comorbidity Index:
 - Mean scores were 0.82 (SD 1.51) for DM patients versus 0.44 (SD 1.19) for MCs
 - Percent of patients with values >1 was 19.1% for DM patients versus 9.7% for MCs
- Overall, compared with MCs:
 - **Conditions:** 160 out of 283 AHRQ categories had significantly higher pre-index use in DM patients (top categories by difference shown in Figure 1). One category had significantly greater use in MCs
 - **Costs:** 45 AHRQ categories had significantly different pre-index costs between cohorts (40 higher for DM; 5 higher for MCs). The top AHRQ categories by cost difference are shown in Figure 2
 - **Services:** 77 AHRQ categories had a significantly different number of pre-index services between cohorts (76 higher for DM; 1 higher for MCs). The top AHRQ categories by difference in the number of services are shown in Figure 3
- DM patients' annual PMPY total cost, days of service, and PMPY services requiring healthcare are shown in Table 2

Table 1. Age, US Region, Insurance, and Payer Type Were Similar for DM Patients and MCs

Descriptive Characteristics	DM Patients (N=1,694)
Gender, % female	50.5%
Age, mean (SD) years	43.4 (18.1)
Age, years	
<18	10.8%
≥18 to <35	19.3%
≥35 to <45	18.2%
≥45 to <55	20.1%
≥55 to <65	23.0%
≥65	8.6%
US region	
South	30.5%
Midwest	28.5%
Northeast	20.8%
West	18.5%
Unknown	1.7%

Descriptive Characteristics	DM Patients (N=1,694)
Insurance type	
Preferred provider organization	66.0%
Health maintenance organization	23.0%
Point-of-service plan	5.4%
Consumer-directed healthcare	2.8%
Indemnity/traditional plan	1.6%
Unknown plan	1.2%
Payer Type	
Commercial	60.3%
Self-insured	21.5%
Medicaid	9.0%
Medicare Advantage	5.5%
Medicare Supplemental	2.8%
Unknown line of business	0.9%

There were no significant differences between DM patients and MCs

Sponsorship
This poster is sponsored by Avidity Biosciences

Abbreviations
AHRQ, Agency for Healthcare Research and Quality; CPI, Consumer Price Index; DM, myotonic dystrophy; ICD, International Classification of Disease; MC, matched control; PMPY, per member per year; Rx, prescription; SD, standard deviation.

Prevalence of Healthcare Conditions and Services Used by Patients with Myotonic Dystrophy (DM1 and DM2) Prior to Diagnosis: a Real-World Data Analysis

John W. Day[1], Kathryn A. Munoz[2], Richard A. Brook[3], Bradley McEvoy[2], Mark C. Stahl[2], Kelly DiTrapani[2], Nathan L. Kleinman[3], Chao-Yin Chen[2], Li-Jung Tai[2]

[1]Stanford University Medical Center; [2]Avidity Biosciences, Inc.; [3]Better Health Worldwide. All authors have met authorship criteria.

Figure 1. Prior to Diagnosis, DM Patients had Higher Absolute Differences[a] in Prevalence for 160 AHRQ Categories vs MCs

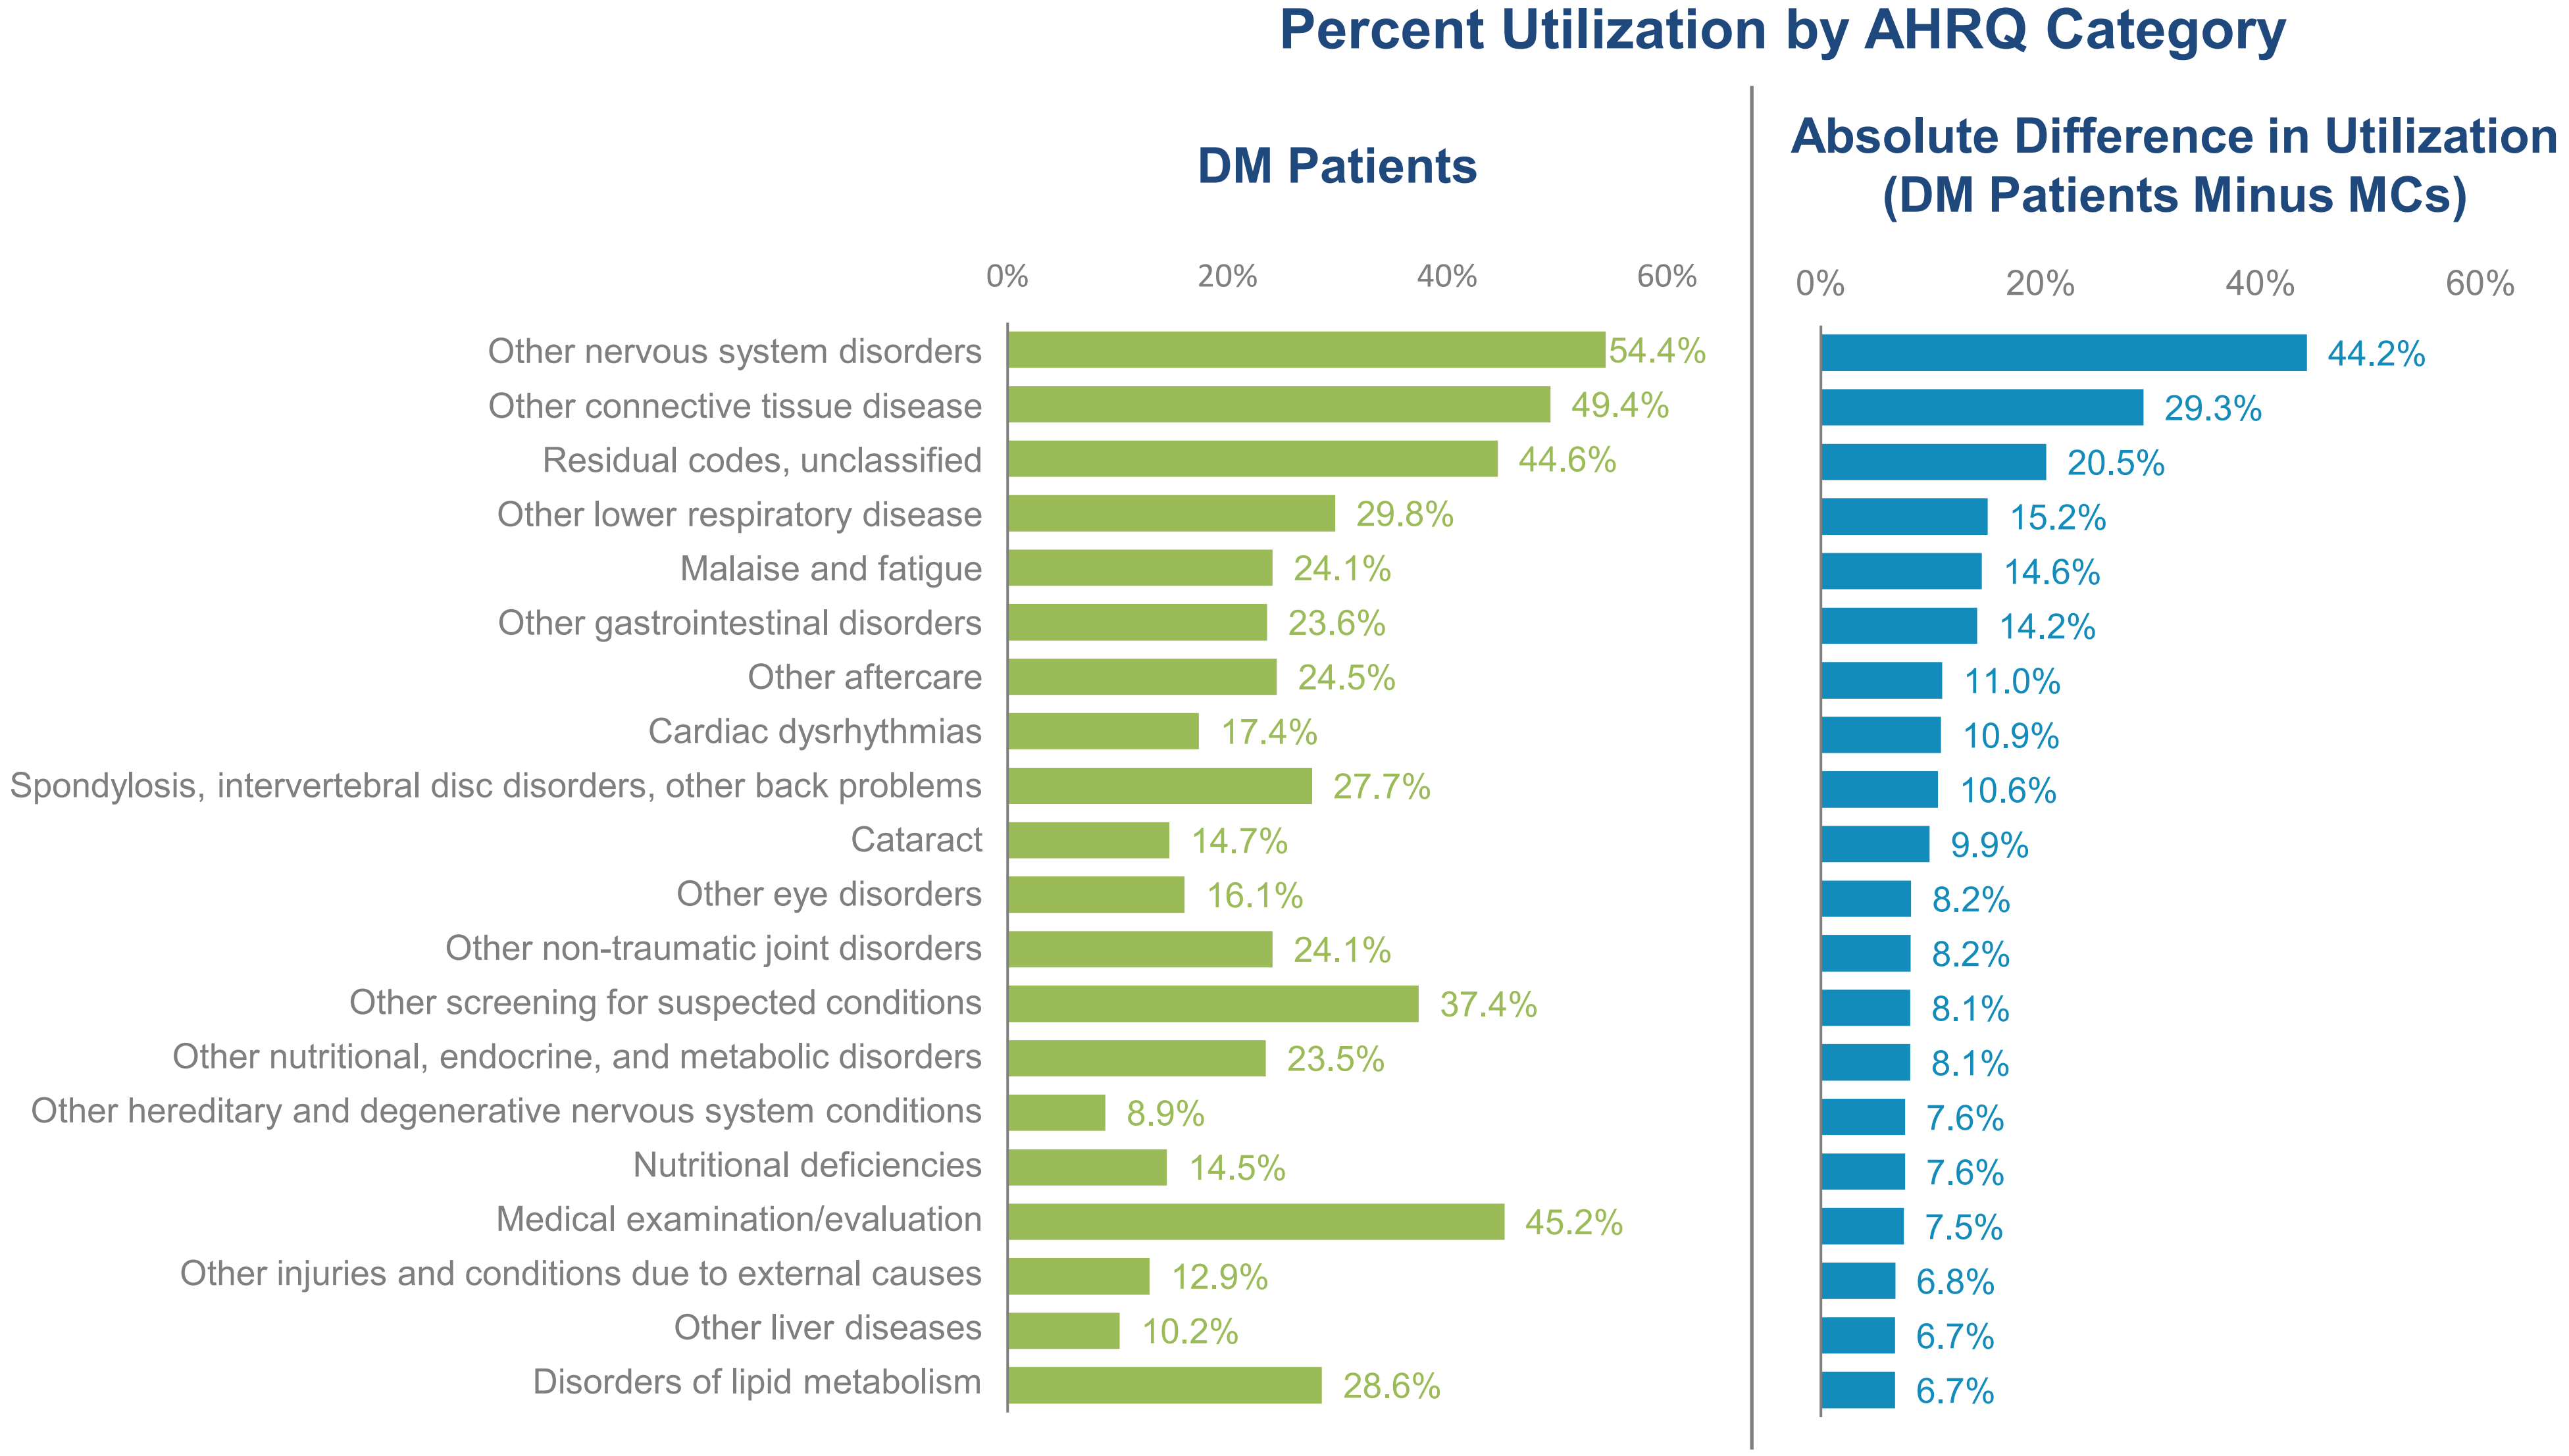
Percent Utilization by AHRQ Category
DM Patients
Absolute Difference in Utilization (DM Patients Minus MCs)
0%
20%
40%
60%
Other nervous system disorders 54.4% 44.2%
Other connective tissue disease 49.4% 29.3%
Residual codes, unclassified 44.6% 20.5%
Other lower respiratory disease 29.8% 15.2%
Malaise and fatigue 24.1% 14.6%
Other gastrointestinal disorders 23.6% 14.2%
Other aftercare 24.5% 11.0%
Cardiac dysrhythmias 17.4% 10.9%
Spondylosis, intervertebral disc disorders, other back problems 27.7% 10.6%
Cataract 14.7% 9.9%
Other eye disorders 16.1% 8.2%
Other non-traumatic joint disorders 24.1% 8.2%
Other screening for suspected conditions 37.4% 8.1%
Other nutritional, endocrine, and metabolic disorders 23.5% 8.1%
Other hereditary and degenerative nervous system conditions 8.9% 7.6%
Nutritional deficiencies 14.5% 7.6%
Medical examination/evaluation 45.2% 7.5%
Other injuries and conditions due to external causes 12.9% 6.8%
Other liver diseases 10.2% 6.7%
Disorders of lipid metabolism 28.6% 6.7%

[a]Absolute difference=DM cohort minus MC cohort
All differences between cohorts $p<0.000177$

Figure 2. Prior to Diagnosis, DM Patients had Higher Costs in AHRQ Categories of “Other Nervous System Disorders”, “Respiratory Failure, Insufficiency, Arrest”, “Septicemia”, “Other Connective Tissue Disorders”, and “Other Gastrointestinal Disorders” versus MCs

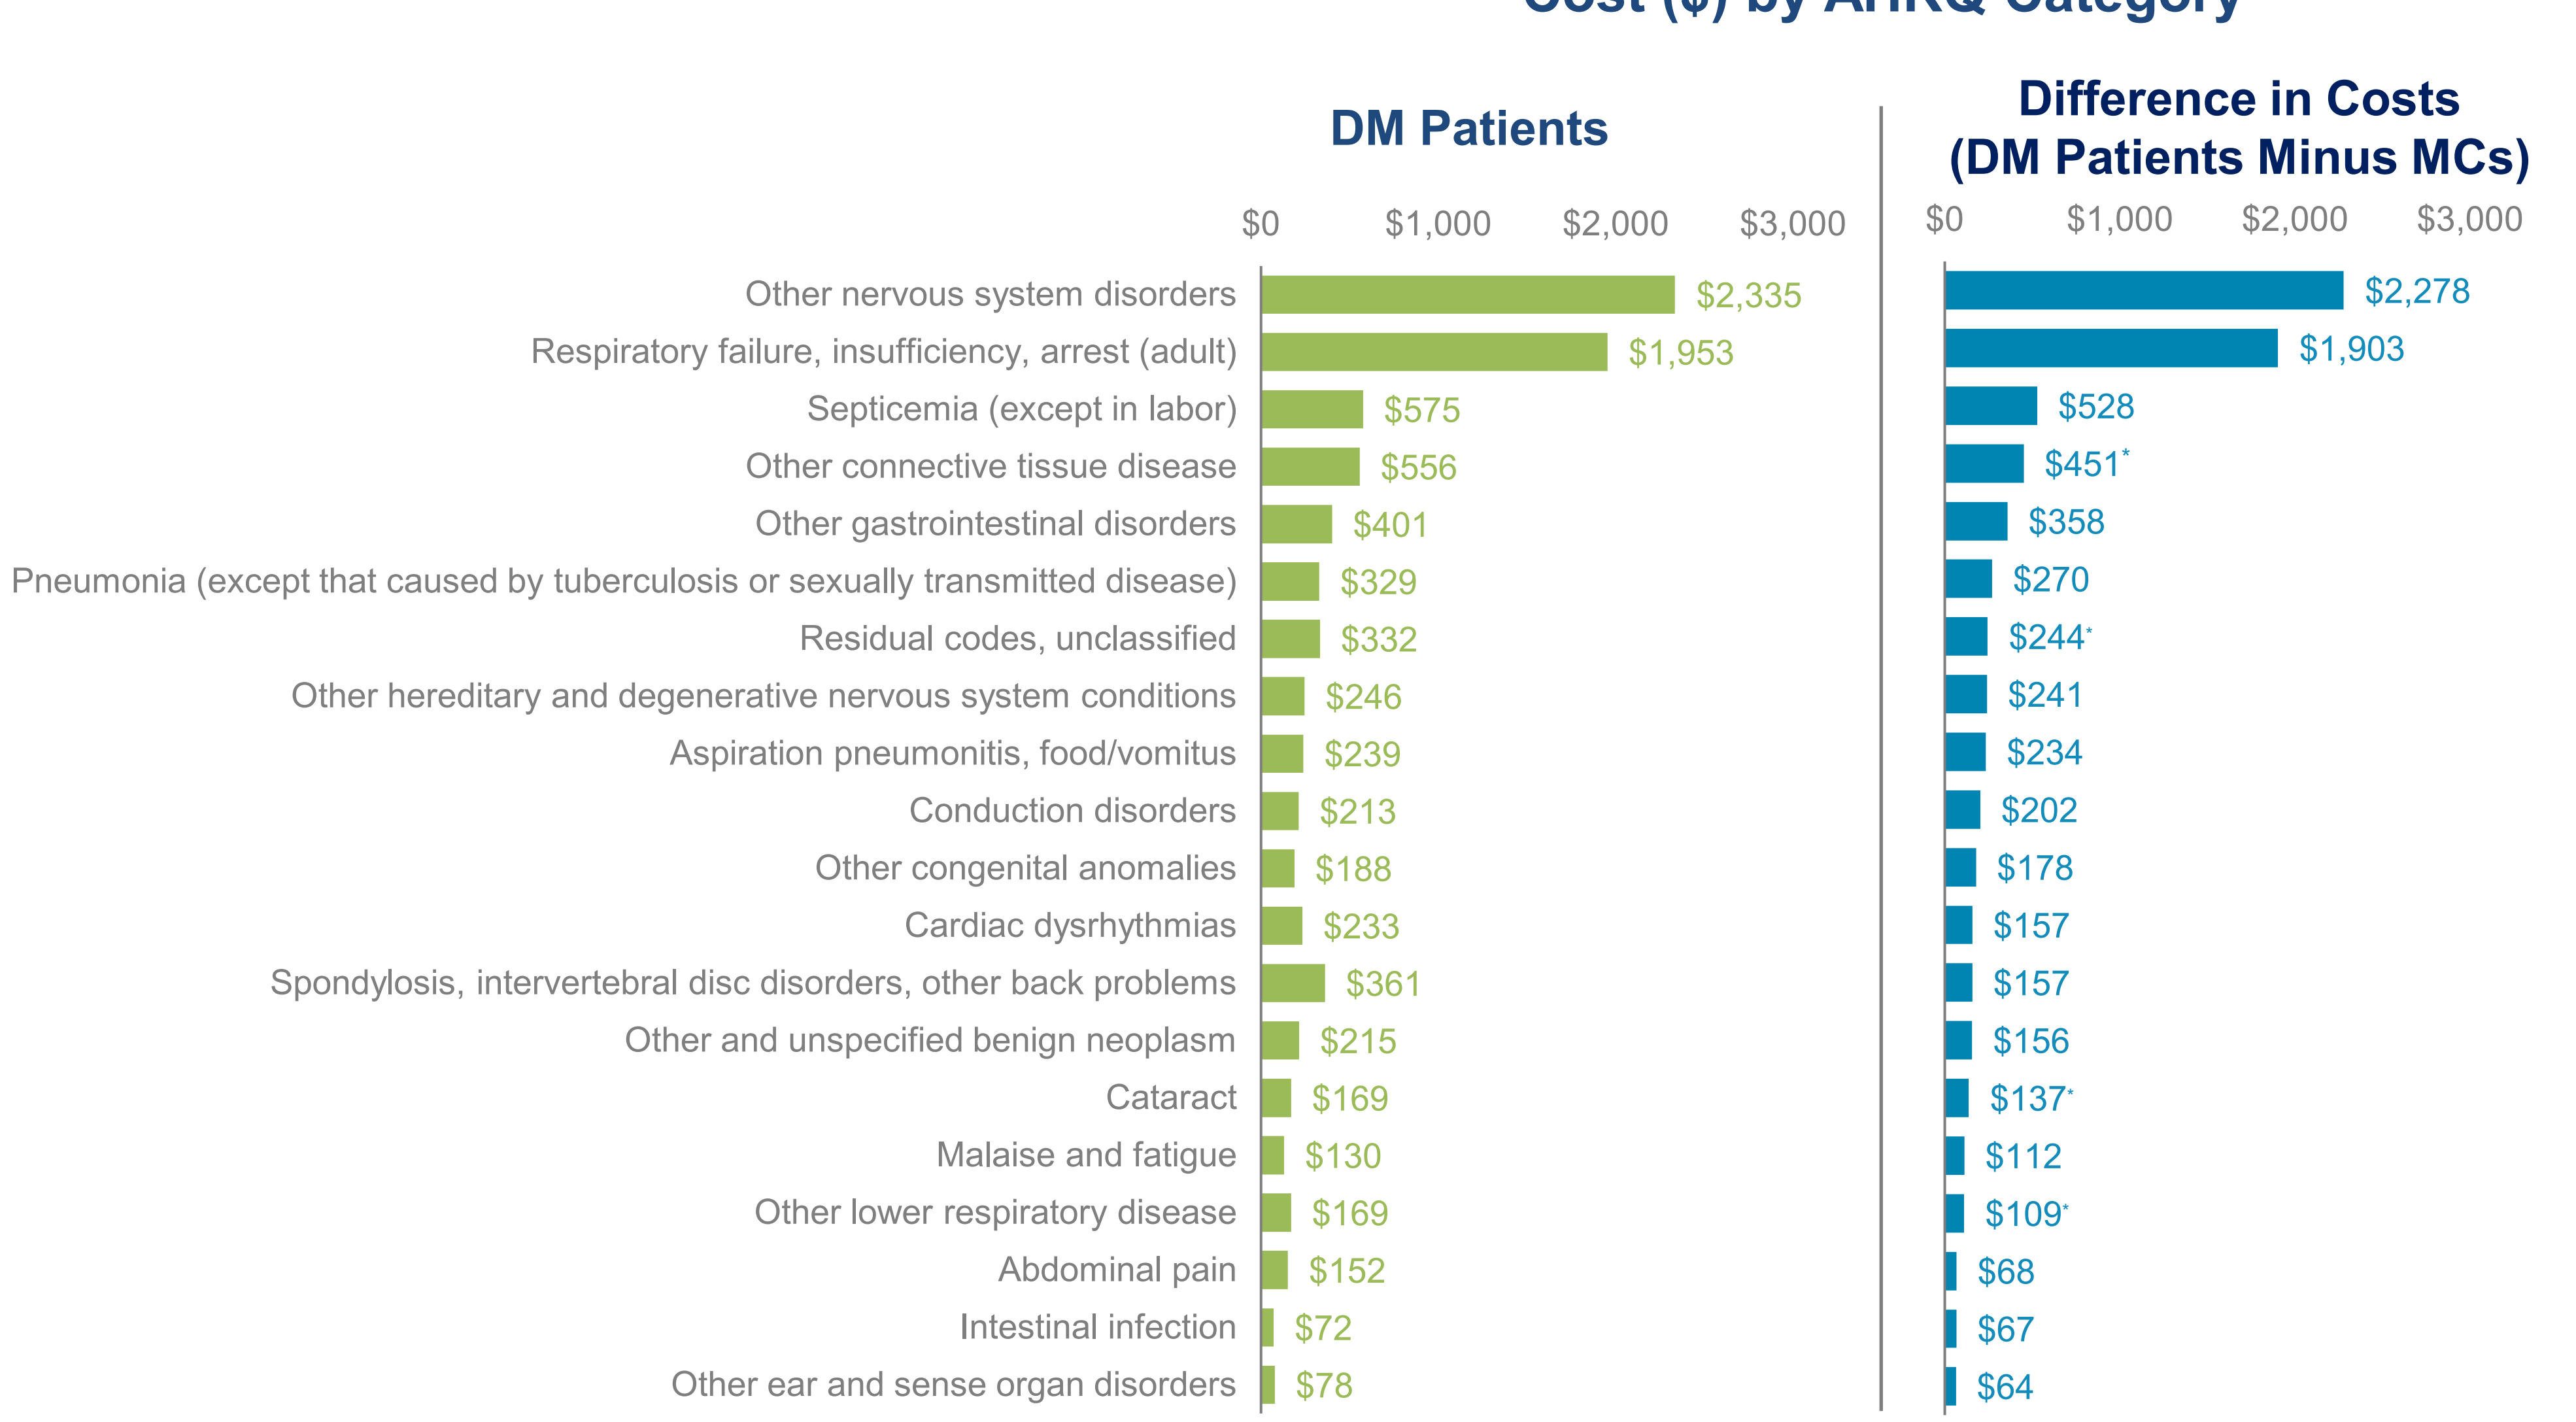
Cost ($) by AHRQ Category
DM Patients
Difference in Costs (DM Patients Minus MCs)
$0
$1,000
$2,000
$3,000
Other nervous system disorders $2,335 $2,278
Respiratory failure, insufficiency, arrest (adult) $1,953 $1,903
Septicemia (except in labor) $575 $528
Other connective tissue disease $556 $451*
Other gastrointestinal disorders $401 $358
Pneumonia (except that caused by tuberculosis or sexually transmitted disease) $329 $270
Residual codes, unclassified $332 $244*
Other hereditary and degenerative nervous system conditions $246 $241
Aspiration pneumonitis, food/vomitus $239 $234
Conduction disorders $213 $202
Other congenital anomalies $188 $178
Cardiac dysrhythmias $233 $157
Spondylosis, intervertebral disc disorders, other back problems $361 $157
Other and unspecified benign neoplasm $215 $156
Cataract $169 $137*
Malaise and fatigue $130 $112
Other lower respiratory disease $169 $109*
Abdominal pain $152 $68
Intestinal infection $72 $67
Other ear and sense organ disorders $78 $64

All $p<0.05$.
*$p<0.000177$ (threshold for multiple comparisons [0.05/283 comparisons])

Sponsorship
This poster is sponsored by Avidity Biosciences

Abbreviations
AHRQ, Agency for Healthcare Research and Quality; CPI, Consumer Price Index; DM, myotonic dystrophy; ICD, International Classification of Disease; MC, matched control; PMPY, per member per year; Rx, prescription; SD, standard deviation.

Prevalence of Healthcare Conditions and Services Used by Patients with Myotonic Dystrophy (DM1 and DM2) Prior to Diagnosis: a Real-World Data Analysis

John W. Day[1], Kathryn A. Munoz[2], Richard A. Brook[3], Bradley McEvoy[2], Mark C. Stahl[2], Kelly DiTrapani[2], Nathan L. Kleinman[3], Chao-Yin Chen[2], Li-Jung Tai[2]

[1]Stanford University Medical Center; [2]Avidity Biosciences, Inc.; [3]Better Health Worldwide. All authors have met authorship criteria.

Figure 3. Prior to Diagnosis, DM Patients had More Services Billed for “Other Nervous System Disorders”, “Other Connective Tissue Disease”, and “Respiratory Failure, Insufficiency, Arrest” Versus MCs

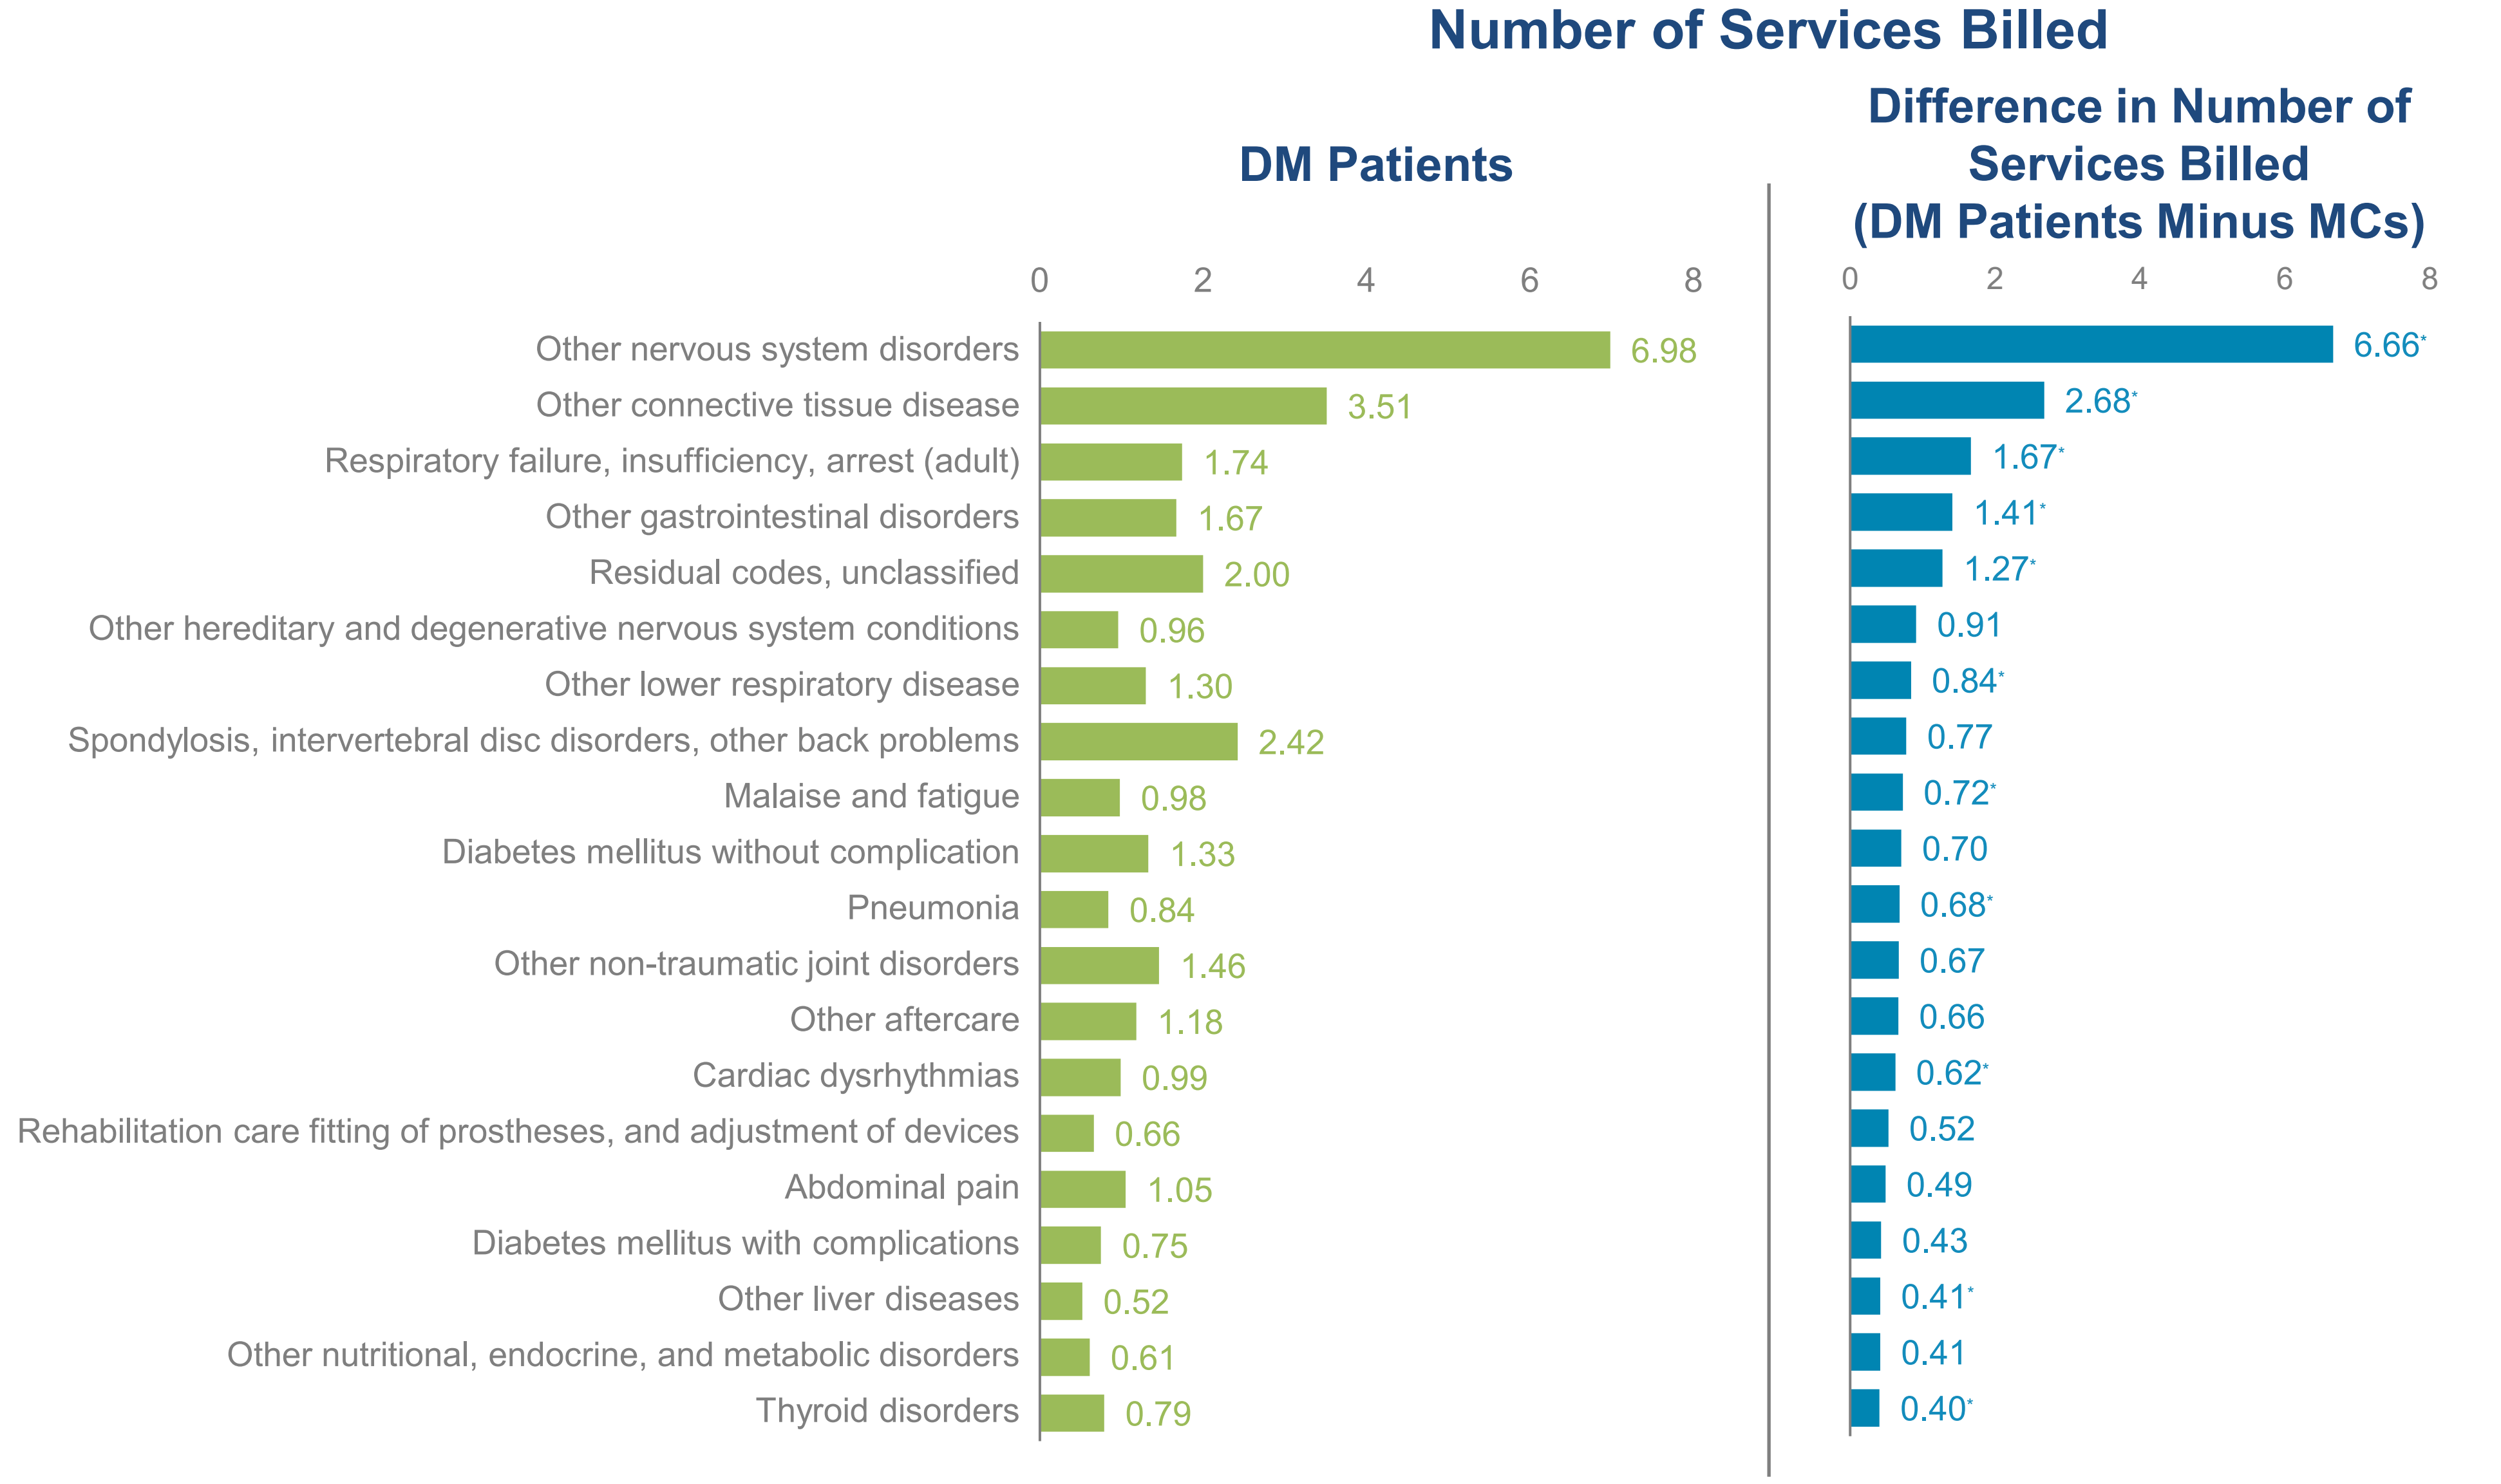
Number of Services Billed
DM Patients
Difference in Number of Services Billed (DM Patients Minus MCs)
0
2
4
6
8
Other nervous system disorders 6.98 6.66*
Other connective tissue disease 3.51 2.68*
Respiratory failure, insufficiency, arrest (adult) 1.74 1.67*
Other gastrointestinal disorders 1.67 1.41*
Residual codes, unclassified 2.00 1.27*
Other hereditary and degenerative nervous system conditions 0.96 0.91
Other lower respiratory disease 1.30 0.84*
Spondylosis, intervertebral disc disorders, other back problems 2.42 0.77
Malaise and fatigue 0.98 0.72*
Diabetes mellitus without complication 1.33 0.70
Pneumonia 0.84 0.68*
Other non-traumatic joint disorders 1.46 0.67
Other aftercare 1.18 0.66
Cardiac dysrhythmias 0.99 0.62*
Rehabilitation care fitting of prostheses, and adjustment of devices 0.66 0.52
Abdominal pain 1.05 0.49
Diabetes mellitus with complications 0.75 0.43
Other liver diseases 0.52 0.41*
Other nutritional, endocrine, and metabolic disorders 0.61 0.41
Thyroid disorders 0.79 0.40*

All $p<0.05$.
*$p<0.000177$ (threshold for multiple comparisons [0.05/283 comparisons])

Table 2. DM Patients’ Annual PMPY Total Cost, Days of Service, and PMPY Services Requiring Healthcare

Category		Mean (SD)		Difference (DM minus MCs)*
		DM patients	MCs	
PMPY Total Cost	Medical	\$18,239 (\$62,654)	\$5,609 (\$23,972)	\$12,630
	Rx	\$3,039 (\$13,883)	\$1,478 (\$6,490)	\$1,561
Days of Service Requiring Healthcare	Medical	20.32 (27.84)	9.32 (15.45)	11.00
	Rx	13.66 (15.98)	8.81 (12.53)	4.85
PMPY Services Requiring Healthcare	Medical	68.32 (99.01)	30.76 (57.49)	37.56
	Rx	20.07 (27.95)	13.33 (22.12)	6.74

*$p<0.0001$

Conclusions

- The journey to a DM diagnosis encounters a higher prevalence of services and costs associated with care of various nervous system disorders, connective tissue disorders, and respiratory conditions, as indicated by analysis of AHRQ and PMPY, respectively
 - This is consistent with the notion that the DM diagnosis is complex, and that symptom management on the journey to diagnosis contributes to the cost and burden of DM to patients and society
 - Identifying patterns of care prior to diagnosis of DM may support earlier diagnosis
- Based on the high unmet need, Avidity Biosciences is investigating AOC 1001 for the potential treatment of myotonic dystrophy type 1
- Future research is needed to investigate changes in AHRQ condition categories that occur after diagnosis and the patterns of concomitant therapy preceding diagnosis

Sponsorship
This poster is sponsored by Avidity Biosciences

Abbreviations
AHRQ, Agency for Healthcare Research and Quality; CPI, Consumer Price Index; DM, myotonic dystrophy; ICD, International Classification of Disease; MC, matched control; PMPY, per member per year; Rx, prescription; SD, standard deviation.