AOC 1001 Demonstrates DMPK Reduction and Spliceopathy Improvement in a Phase 1/2 Study in Myotonic Dystrophy Type 1 (DM1) (MARINA™)

Yiming Zhu, Tanya Kwan, Qingying Meng, Li-Jung Tai, Michelle Lee, Husam Younis, Art Levin, Mike Flanagan

Avidity Biosciences, Inc., San Diego, CA 92121

Background

Myotonic dystrophy type 1 (DM1) is a rare, autosomal dominant, progressive neuromuscular disease with no US Food and Drug Administration (FDA)-approved therapies. The cause of DM1 is a mutation in myotonic dystrophy protein kinase (*DMPK*) messenger ribonucleic acid (mRNA) that contains expanded (>50) CUG repeats (Figure 1a).[1–5] Mutant *DMPK* is a toxic gain-of-function mRNA that leads to nuclear retention (nuclear foci) and sequestration of splicing factors (e.g., muscleblind-like [MBNL]) resulting in global splicing dysregulation.[6]

Given the challenges with delivery of oligonucleotides to muscle, we utilized transferrin receptor 1 (TfR1)-mediated endocytosis to deliver siRNA to skeletal muscle and heart. Thus, AOC 1001 is an antibody oligonucleotide conjugate (AOC™) comprised of an siRNA targeting *DMPK* mRNA (siDMPK) conjugated to a humanized monoclonal antibody (mAb) targeting human TfR1 that is designed to reduce *DMPK* mRNA in muscle tissue and subsequently correct splicing events that are responsible for DM1 pathogenesis (Figure 1b).

Figure 1: Avidity's Approach to Targeting DM1

1a. Mechanism of Disease | **1b. Potential Therapeutic Approach**

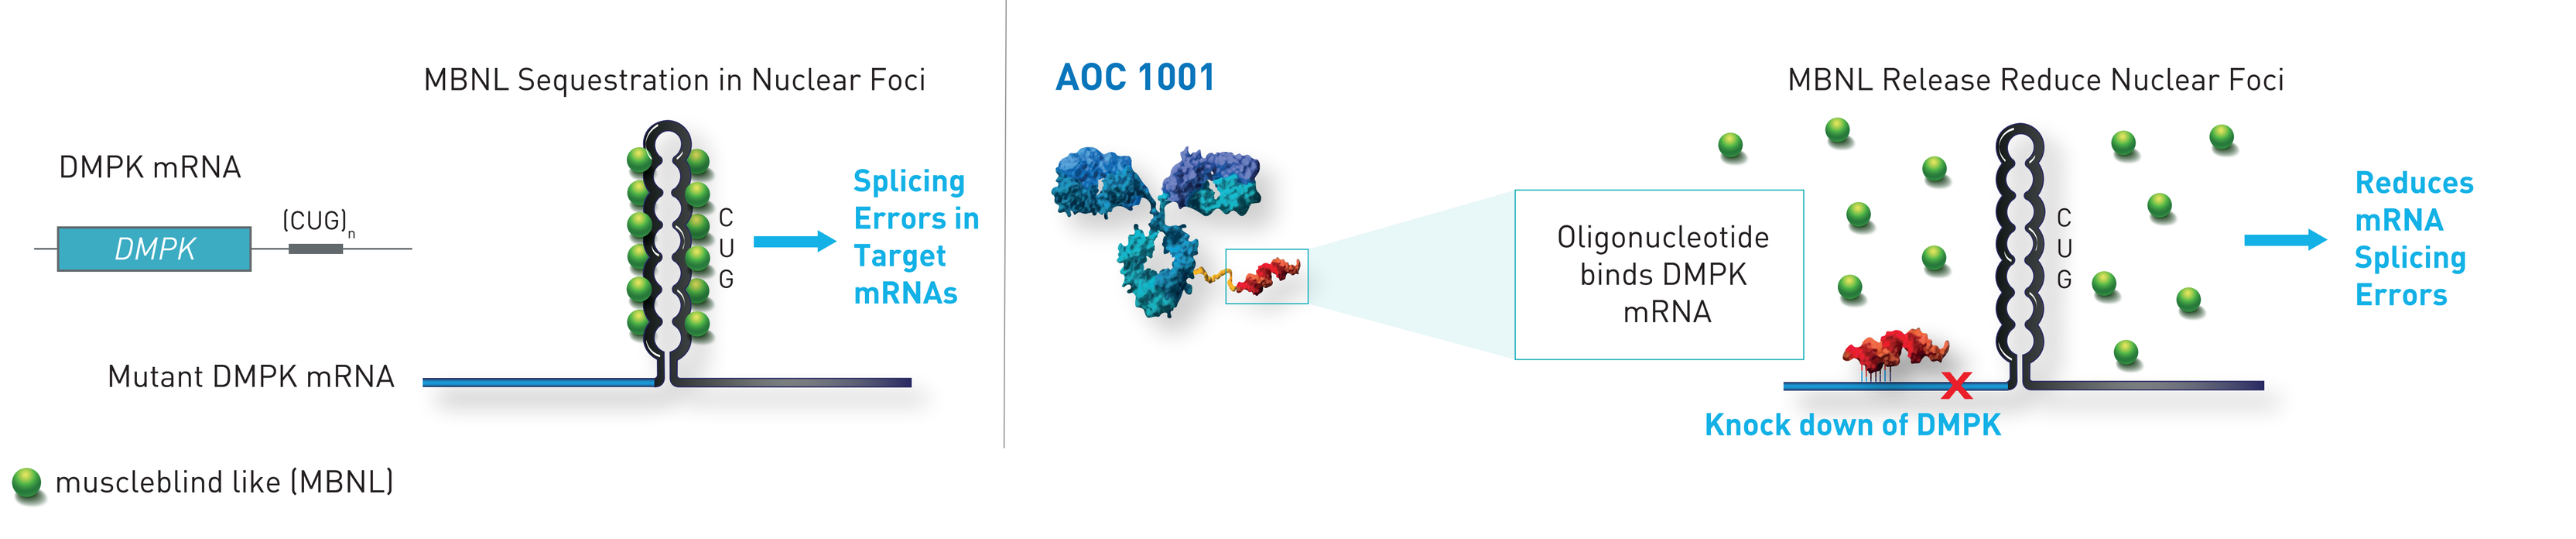
MBNL Sequestration in Nuclear Foci
DMPK mRNA
DMPK
(CUG)n
C U G
Splicing Errors in Target mRNAs
Mutant DMPK mRNA
muscleblind like (MBNL)
AOC 1001
MBNL Release Reduce Nuclear Foci
Oligonucleotide binds DMPK mRNA
Reduces mRNA Splicing Errors
Knock down of DMPK

Avidity's Approach to Treating DM1

- The activity of AOC 1001 was evaluated *in vitro* in DM1 patient myotubes and in a clinical trial of patients with DM1.
- DM1 patient myotubes were transfected with siDMPK. Levels of *DMPK* mRNA and splicing events (RNA sequencing) as well as nuclear foci (immunohistochemistry) were evaluated.
- MARINA™ is a Phase 1/2 study (NCT05027269) to evaluate the safety, tolerability, pharmacokinetics, and pharmacodynamics of AOC 1001 in adult patients with DM1 (Figure 2)*. Pre- and 6 weeks post-dose muscle biopsy samples were evaluated for *DMPK* mRNA expression and splicing changes by qPCR and RNA sequencing, respectively.[†]

Figure 2: Study Design of MARINA™ to Evaluate AOC 1001 in DM1

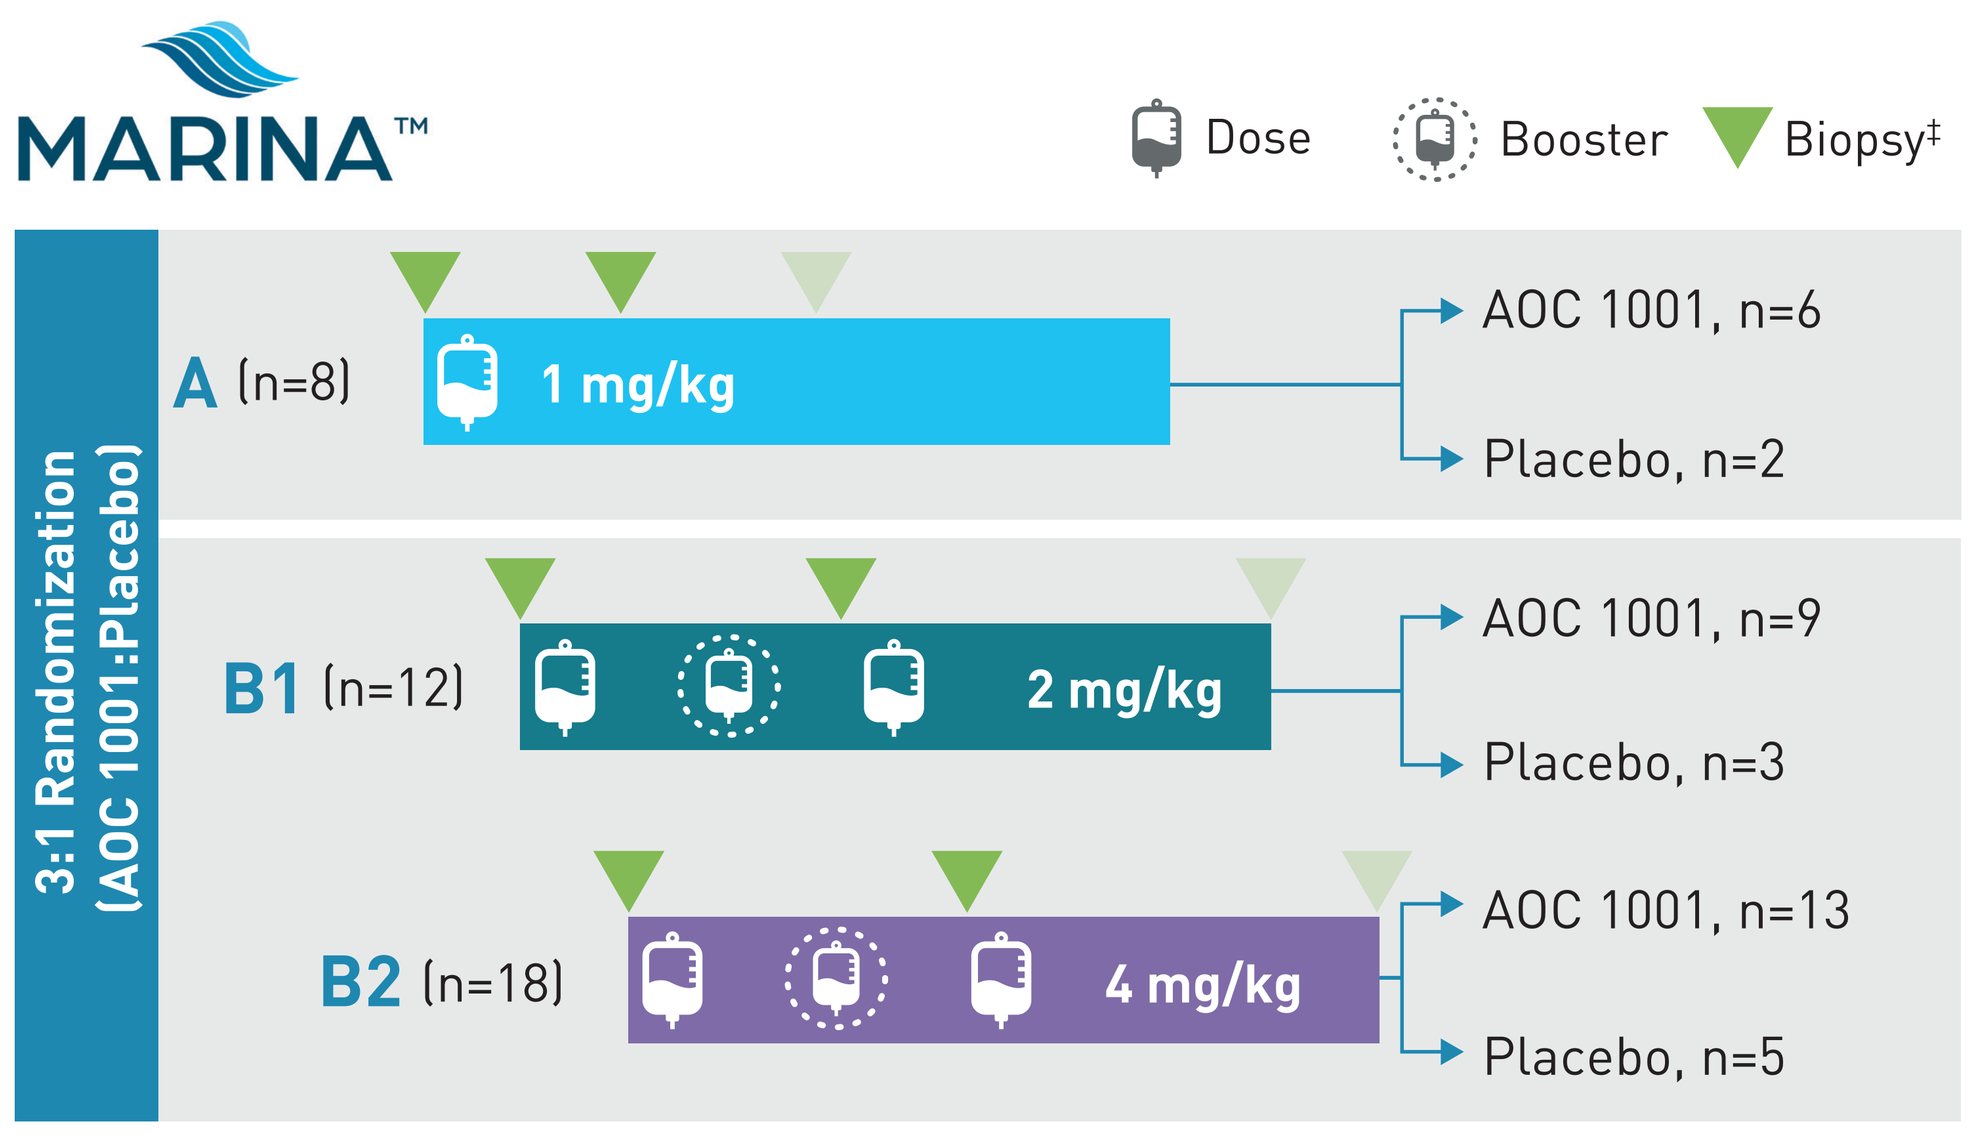
MARINA™
Dose
Booster
Biopsy‡
3:1 Randomization (AOC 1001:Placebo)
A (n=8)
1 mg/kg
AOC 1001, n=6
Placebo, n=2
B1 (n=12)
2 mg/kg
AOC 1001, n=9
Placebo, n=3
B2 (n=18)
4 mg/kg
AOC 1001, n=13
Placebo, n=5

- N=38 patients aged between 18 and 65 years
- **Part A** receives single intravenous (IV) dose
- **Part B** receives multi-ascending IV doses
 - Quarterly doses + one booster after first 6 weeks
- 6-month treatment and observation duration after the first 6 weeks

*In May 2023, the FDA eased the partial clinical hold placed in September 2022 to allow a number of current participants to be dose-escalated to 4 mg/kg of AOC 1001 and new participant enrollment at 2 mg/kg of AOC 1001.
†Includes biomarker data as of April 24, 2023.
‡Biopsies highlighted in dark green were analyzed for this presentation.

Results

Figure 3: siDMPK Reduces *DMPK* mRNA Expression and Improves Spliceopathy and Nuclei Foci in DM1 Myotubes

3a. Knockdown of *DMPK* mRNA Expression | **3b. Reduction of Nuclei Foci** | **3c. Improvement of Spliceopathy**

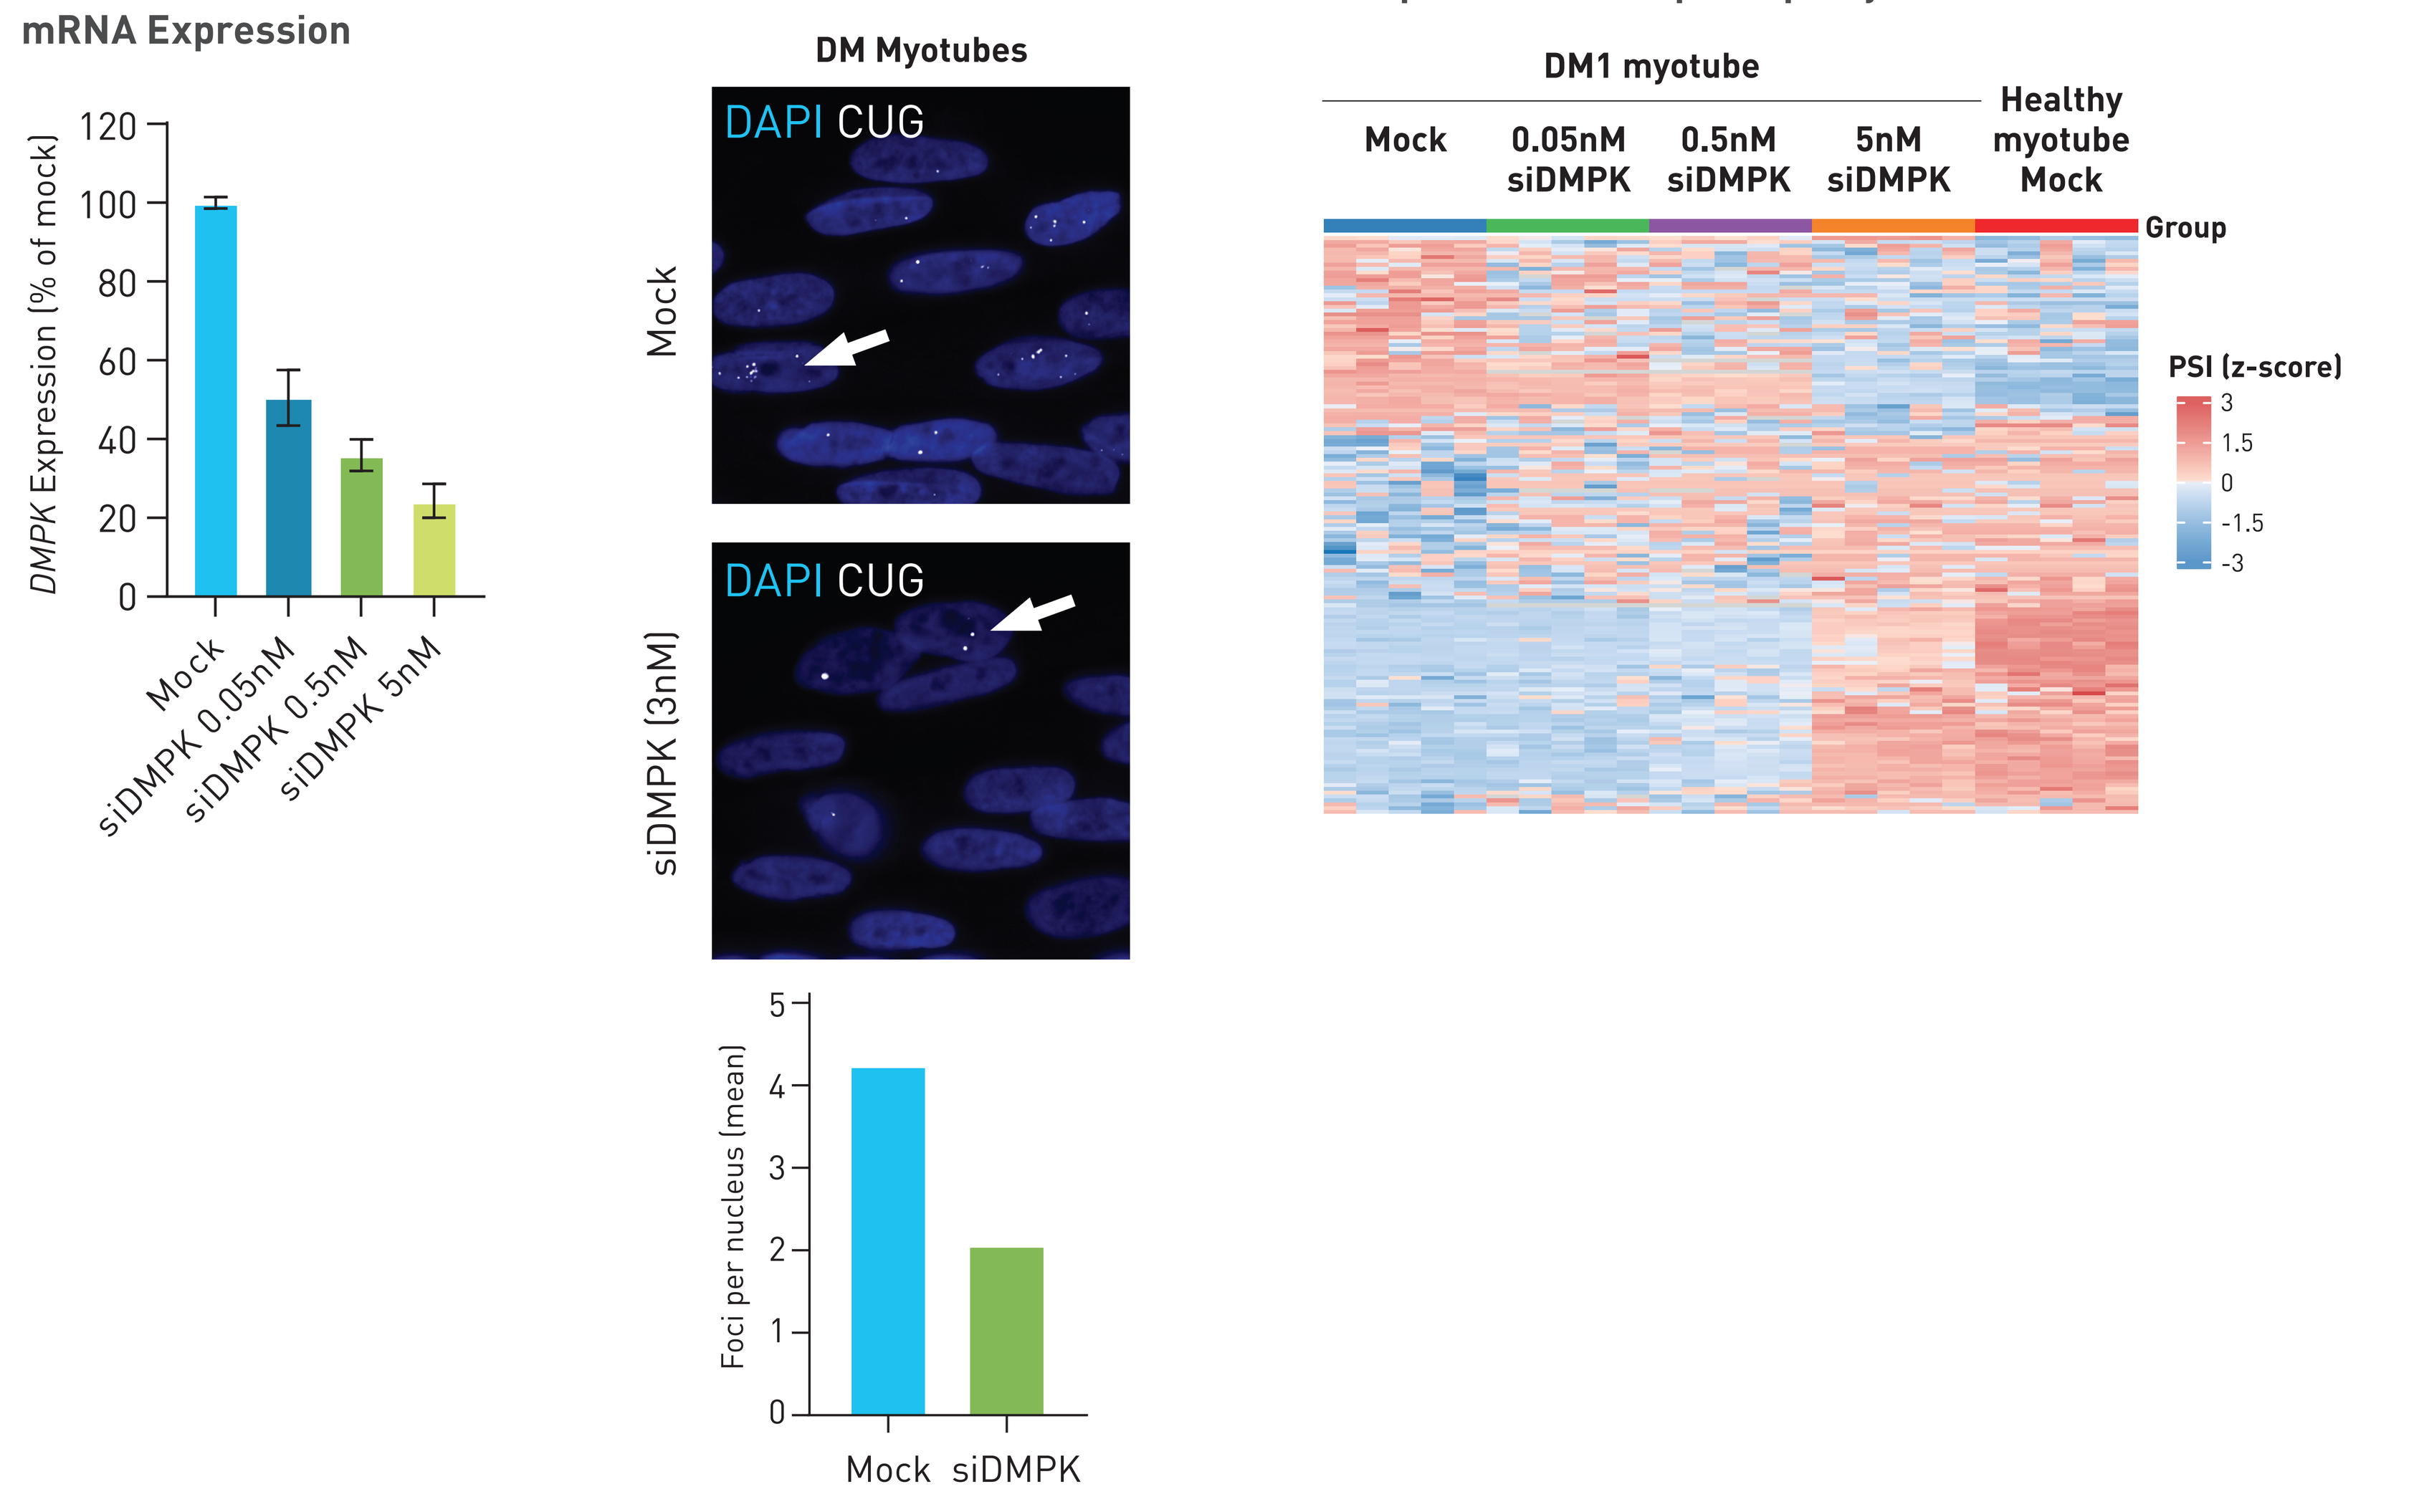
DMPK Expression (% of mock)
Mock
siDMPK 0.05nM
siDMPK 0.5nM
siDMPK 5nM
DM Myotubes
DAPI CUG
Mock
siDMPK (3nM)
Foci per nucleus (mean)
Mock
siDMPK
DM1 myotube
Mock
0.05nM siDMPK
0.5nM siDMPK
5nM siDMPK
Healthy myotube Mock
Group
PSI (z-score)

Figure 3: (a) DMPK expression in DM1 myotubes after treatment with siDMPK (N=5 replicates, mean + standard deviation). (b) Representative images of CUG-containing nuclei foci and mean foci number per nucleus in mock and siDMPK-treated DM1 myotubes. (c) Over 100 mis-spliced events in DM1 myotubes (FDR<0.05 and |ΔPSI|>20 in DM1 myotube versus healthy myotube) were corrected by siDMPK treatment (N=5 replicates). PSI, percent spliced-in.

Results (Continued)

Figure 4: AOC 1001 Reduces *DMPK* mRNA Levels in DM1 Patients

4a. *DMPK* Reduction in all Subjects on Treatment | **4b. Mean Reduction in *DMPK* Expression**

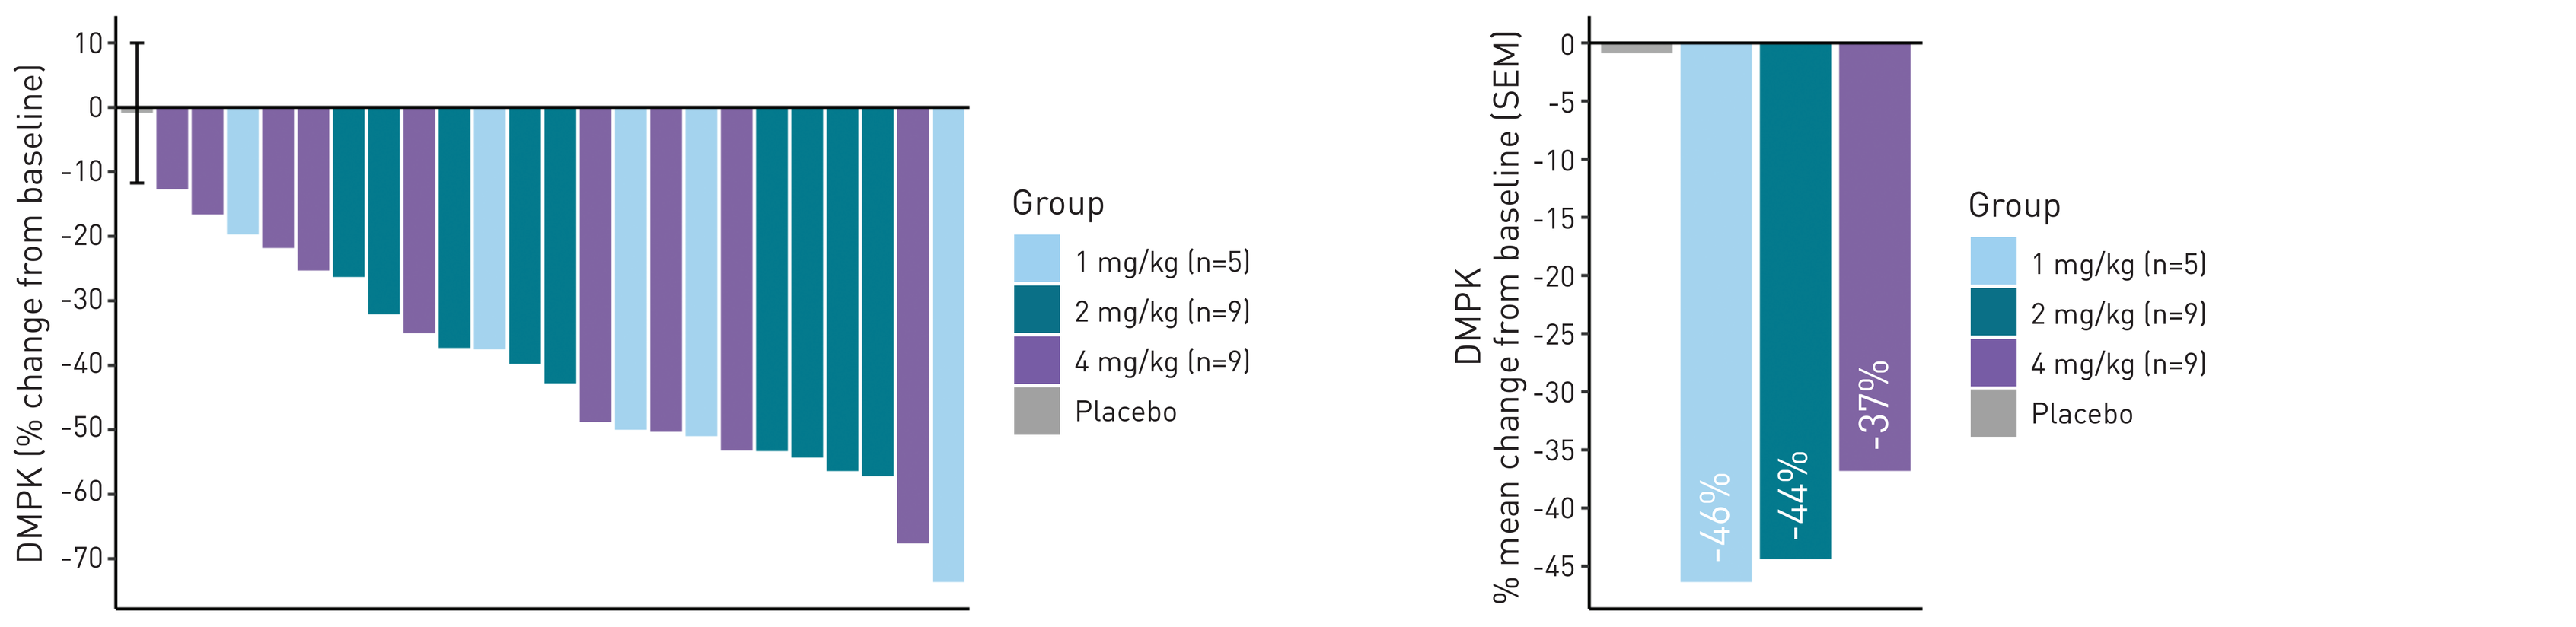
DMPK (% change from baseline)
Group
1 mg/kg (n=5)
2 mg/kg (n=9)
4 mg/kg (n=9)
Placebo
DMPK % mean change from baseline (SEM)
-46%
-44%
-37%

Figure 4: (a) Individual percent change from baseline and (b) mean percent change from baseline of DMPK expression 6 weeks post one dose of 1 mg/kg or two doses of 2 or 4 mg/kg AOC 1001. Placebo group combined and shown as mean and standard error of the mean (SEM).

Figure 5: AOC 1001 Improves Splicing of 22-Gene Panel in DM1 Patients

5a. 22-Gene Splicing Panel Score Change from Baseline in all Subjects on Treatment | **5b. Mean Reduction in 22-Gene Splicing Panel**

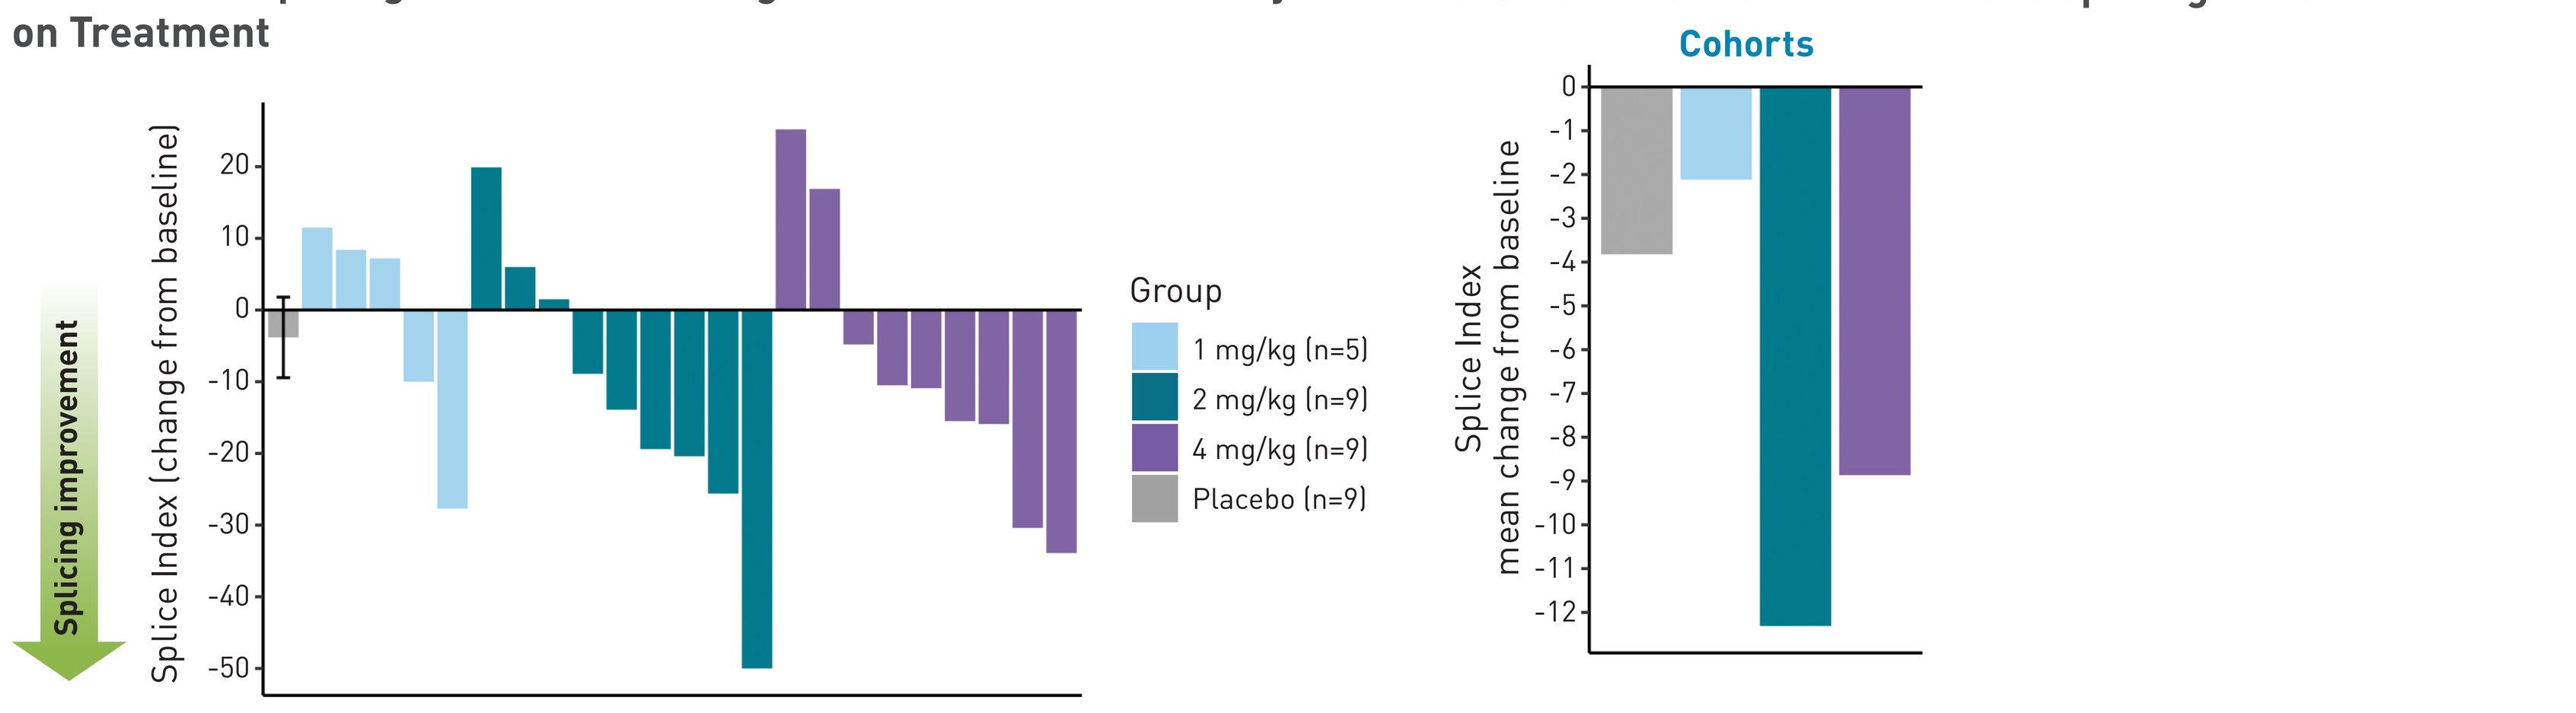
Splicing improvement
Splice Index (change from baseline)
Group
1 mg/kg (n=5)
2 mg/kg (n=9)
4 mg/kg (n=9)
Placebo (n=9)
Cohorts
Splice Index mean change from baseline

Figure 5: Splicing improvements in 22-gene panel demonstrates AOC 1001 is impacting DM1 disease mechanism. (a) Individual change from baseline and (b) mean change from baseline in 22-gene splicing score 6 weeks post one dose of 1 mg/kg or two doses of 2 or 4 mg/kg AOC 1001. Placebo group combined and shown as mean +/- SEM. Splicing measured by targeted RNA sequencing and calculated using published formula.[6] Mean change from baseline is the mean change from baseline score across all matched samples in a cohort.

Figure 6: AOC 1001 Improves Spliceopathy in DM1 Patients

6a. Global Splicing Changes Observed Following AOC 1001 Treatment

6b. Mean Splicing Improvement in Genes Related to Muscle Function

6c. Mean Increase of Correctly Spliced Isoform in Genes Related to Muscle Function

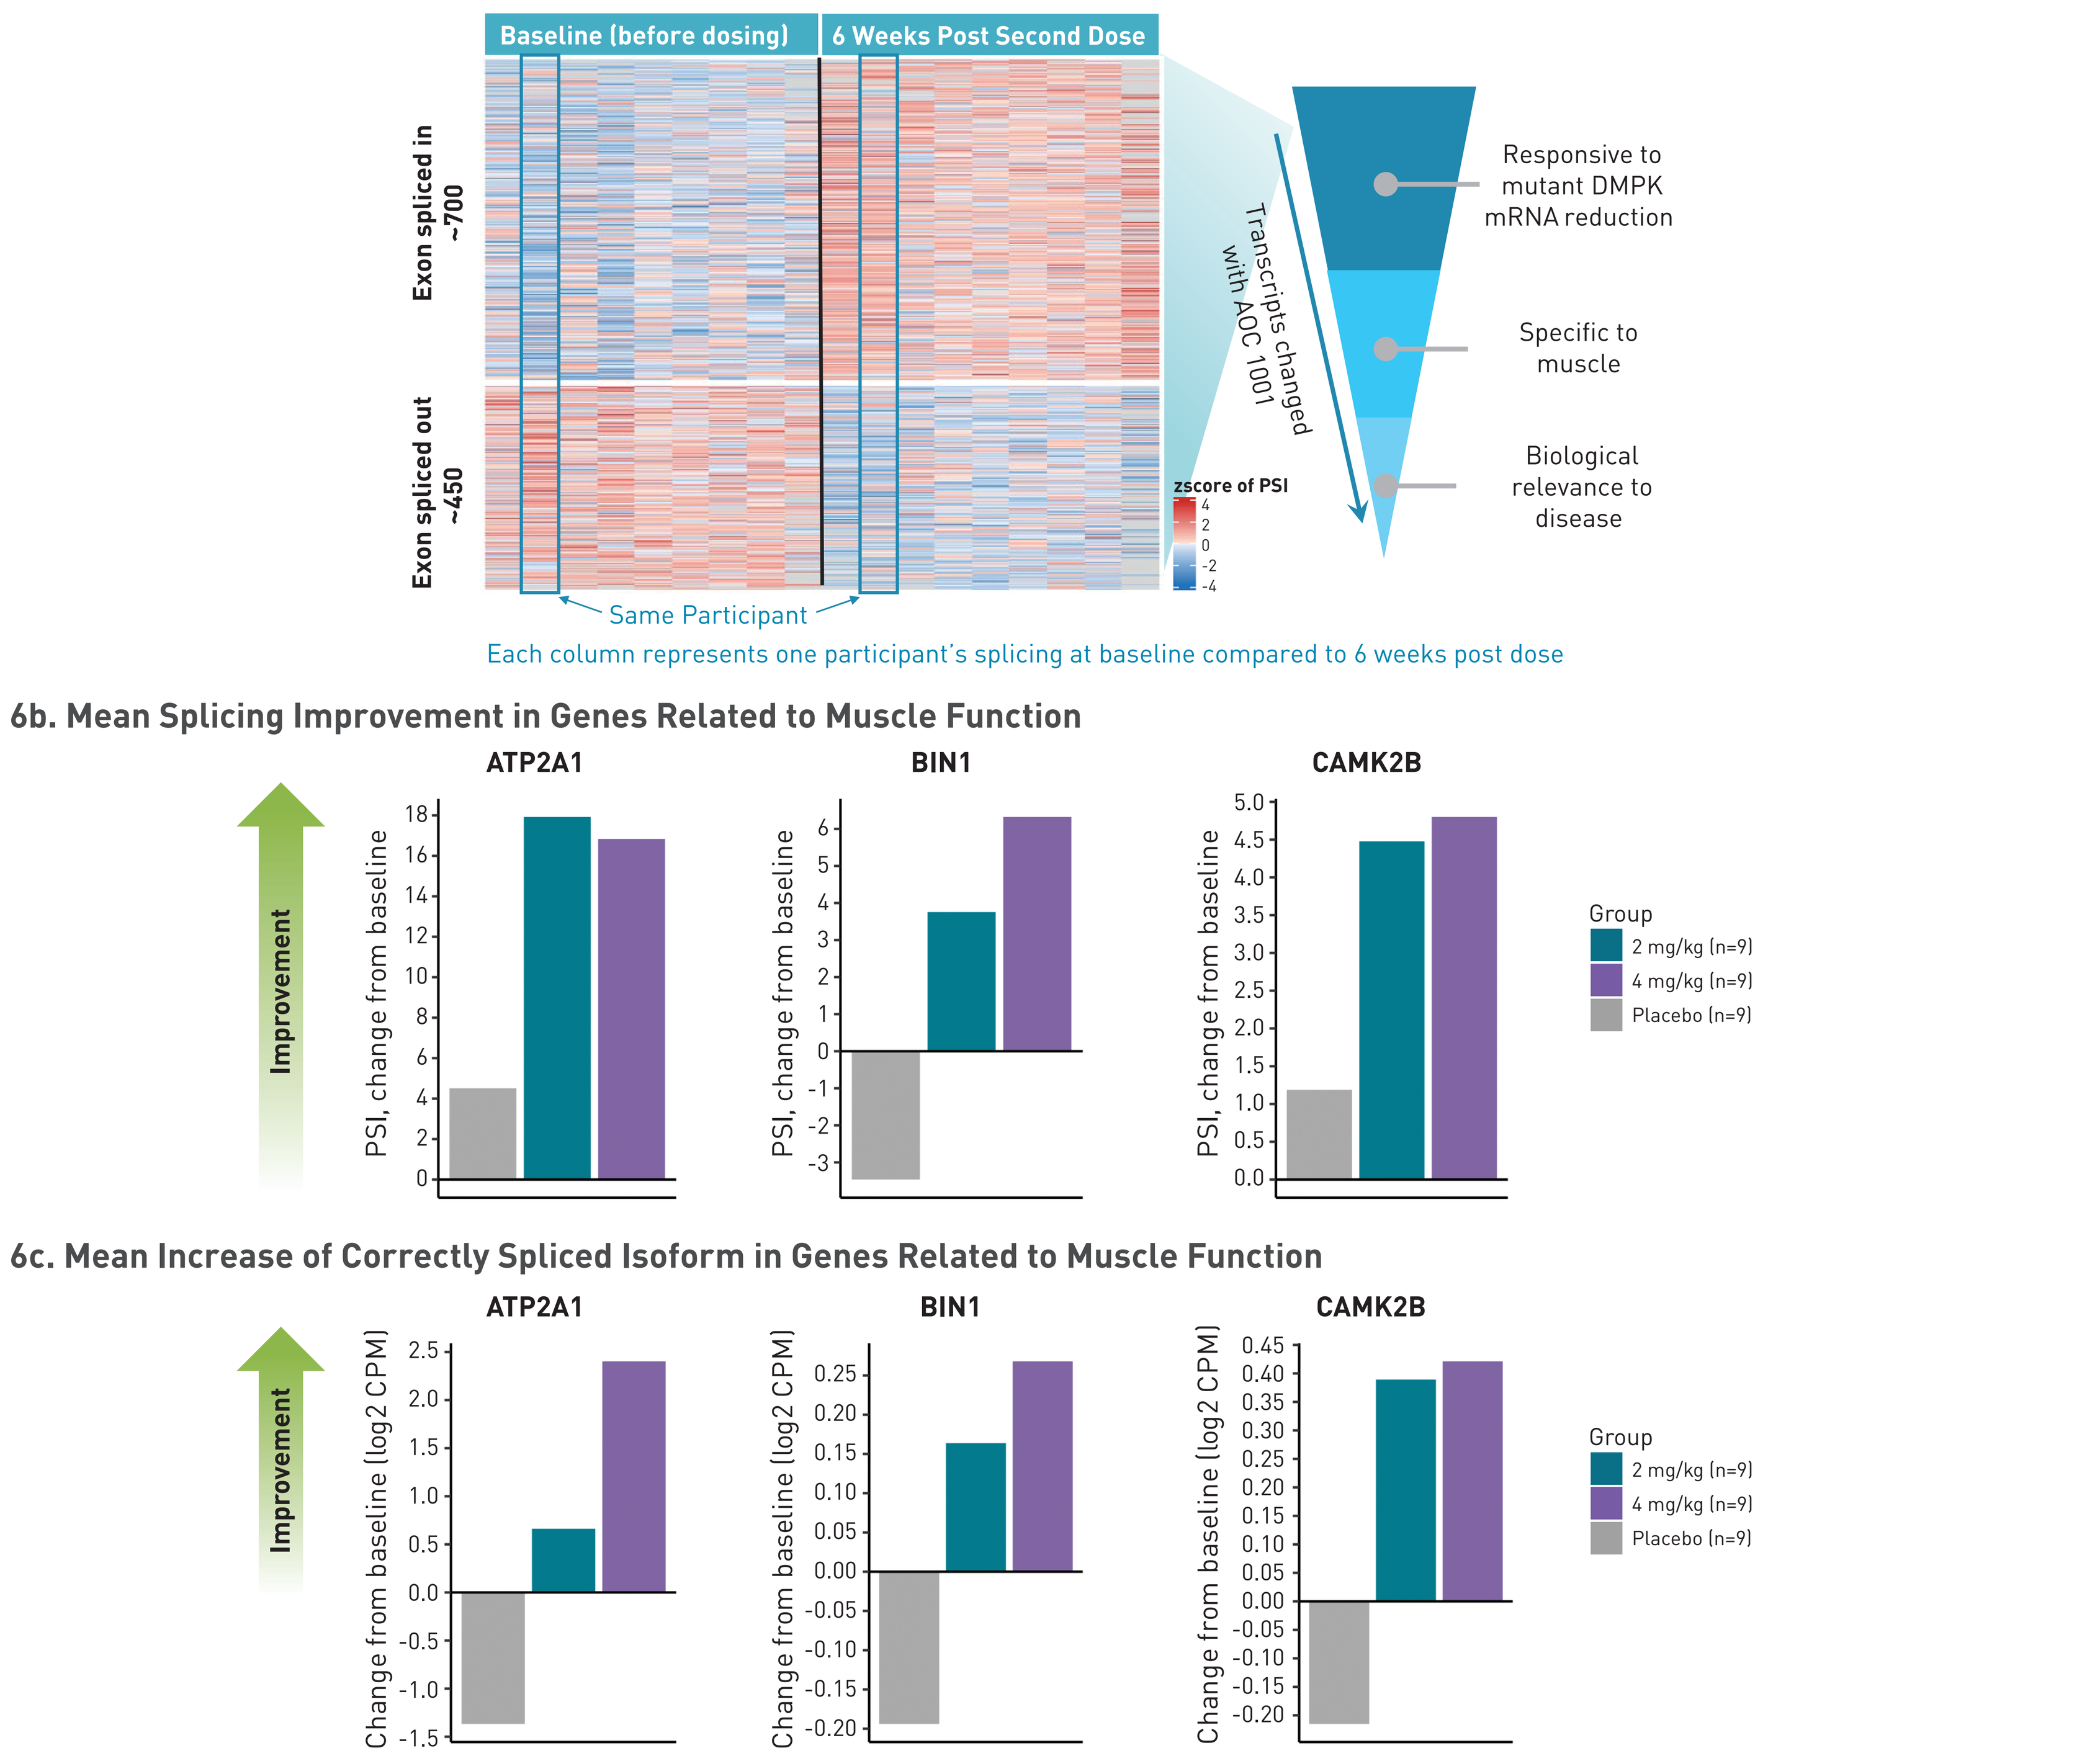
Baseline (before dosing)
6 Weeks Post Second Dose
Exon spliced in ~700
Exon spliced out ~450
zscore of PSI
Transcripts changed with AOC 1001
Responsive to mutant DMPK mRNA reduction
Specific to muscle
Biological relevance to disease
Same Participant
Each column represents one participant's splicing at baseline compared to 6 weeks post dose
Improvement
ATP2A1
BIN1
CAMK2B
PSI, change from baseline
Change from baseline (log2 CPM)
Group
2 mg/kg (n=9)
4 mg/kg (n=9)
Placebo (n=9)

Figure 6: (a) Example of the broad impact AOC 1001 has on global splicing. Bulk RNA sequencing utilized to identify mRNA transcripts with significant splicing changes 6 weeks post second dose of AOC 1001 (2 mg/kg) relative to baseline (p<0.05, mean PSI difference >0.05). (b) PSI change (mean) and (c) expression change of the correctly spliced isoform (mean) of splicing events related to DM1 pathogenesis and muscle function in MARINA participants 6 weeks post second dose of 2 mg/kg or 4mg/kg AOC 1001.

Conclusion

We demonstrate the translation of potent *in vitro* activity to proof-of-mechanism (DMPK reduction and spliceopathy improvement) of AOC 1001 in a first-in-human clinical study in DM1 participants, supporting AOC 1001 as a potential therapy addressing the underlying cause of DM1.

Acknowledgments

Investigators, participants and their families in MARINA™. Association Institut de Myologie for providing control and DM1 cell lines. CHU de Québec-Université Laval, Canada for providing DM1 patient-derived human primary myoblasts.

References

1. LoRusso S, et al. *Neurotherapeutics*. 2018;15(4):872-884.
2. Ashizawa T, et al. *Neurol Clin Pract*. 2018;8(6):507-520.
3. Brook JD, et al. *Cell*. 1992;68(4):799-808.
4. Lin X, et al. *Hum Mol Genet*. 2006;15(13):2087-2097.
5. Lee JE, Cooper TA. *Biochem Soc Trans*. 2009;37(Pt 6):1281-1286.
6. Tanner MK, et al. *Nucleic Acids Res*. 2021;49(4):2240-2254.

Abbreviations

AOC, antibody oligonucleotide conjugate; CPM, count per million; DM1, myotonic dystrophy type 1; DMPK, myotonic dystrophy protein kinase; FDA, US Food and Drug Administration; IV, intravenous; mAb, monoclonal antibody; MBNL, muscleblind like; mRNA, messenger ribonucleic acid; PSI, percent spliced-in; PSO, percent spliced-out; SEM, standard error of the mean; siRNA, small interfering ribonucleic acid; TfR1, transferrin receptor 1.

28th Annual Congress of the World Muscle Society | Charleston, SC | October 3rd–7th, 2023